Presented at 63rd ASH Annual Meeting & Exposition
December 11–14, 2021
Atlanta, GA and Virtual

Posoleucel (ALVR105), an Off-the-Shelf, Multivirus-Specific T-Cell Therapy, for the Prevention of Viral Infections Post-HCT

Results from an Open-Label Cohort of a Phase 2/3 Trial

Sanjeet S. Dadwal[1], Michael Schuster[2], Gary Douglas Myers[3], Keith Boundy[4], Marshelle Warren[5], Elizabeth Stoner[4], Thuy Truong[1], Joshua A. Hill[6]

[1]City of Hope National Medical Center, Duarte, CA; [2]Stony Brook Cancer Center, Stony Brook, NY; [3]Children's Mercy Hospital, Kansas City, MO; [4]AlloVir, Cambridge, MA; [5]Glacier Bio, North Bend, WA; [6]Fred Hutchinson Cancer Research Center, Seattle, WA

Multi-Virus Infections Are Common in Allogeneic HCT Patients and Contribute to Significant Mortality

Incidence of AdV, BKV, CMV, EBV or HHV-6 infection[1] (N=404)

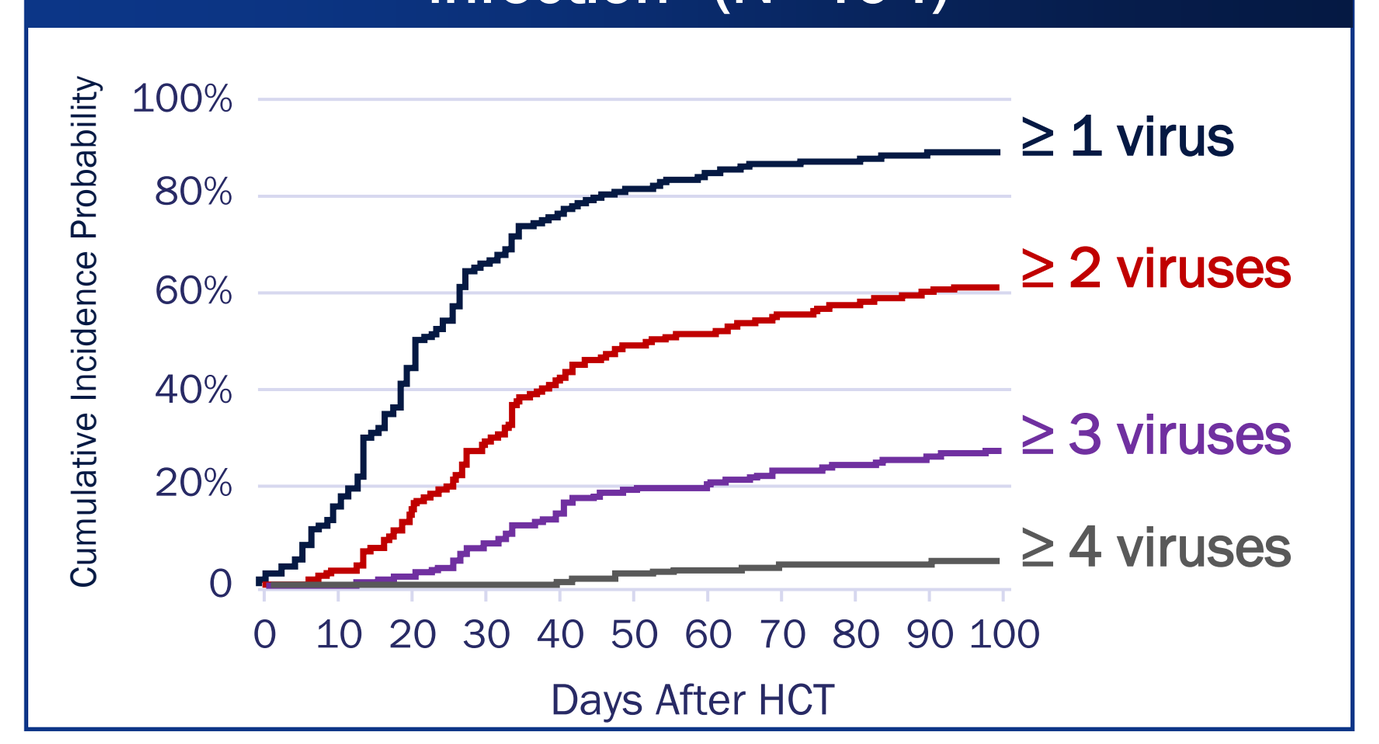
Cumulative Incidence Probability
100%
80%
60%
40%
20%
0
0 10 20 30 40 50 60 70 80 90 100
Days After HCT
≥ 1 virus
≥ 2 viruses
≥ 3 viruses
≥ 4 viruses

Overall survival, number of viruses at the same time[1] (N=358)

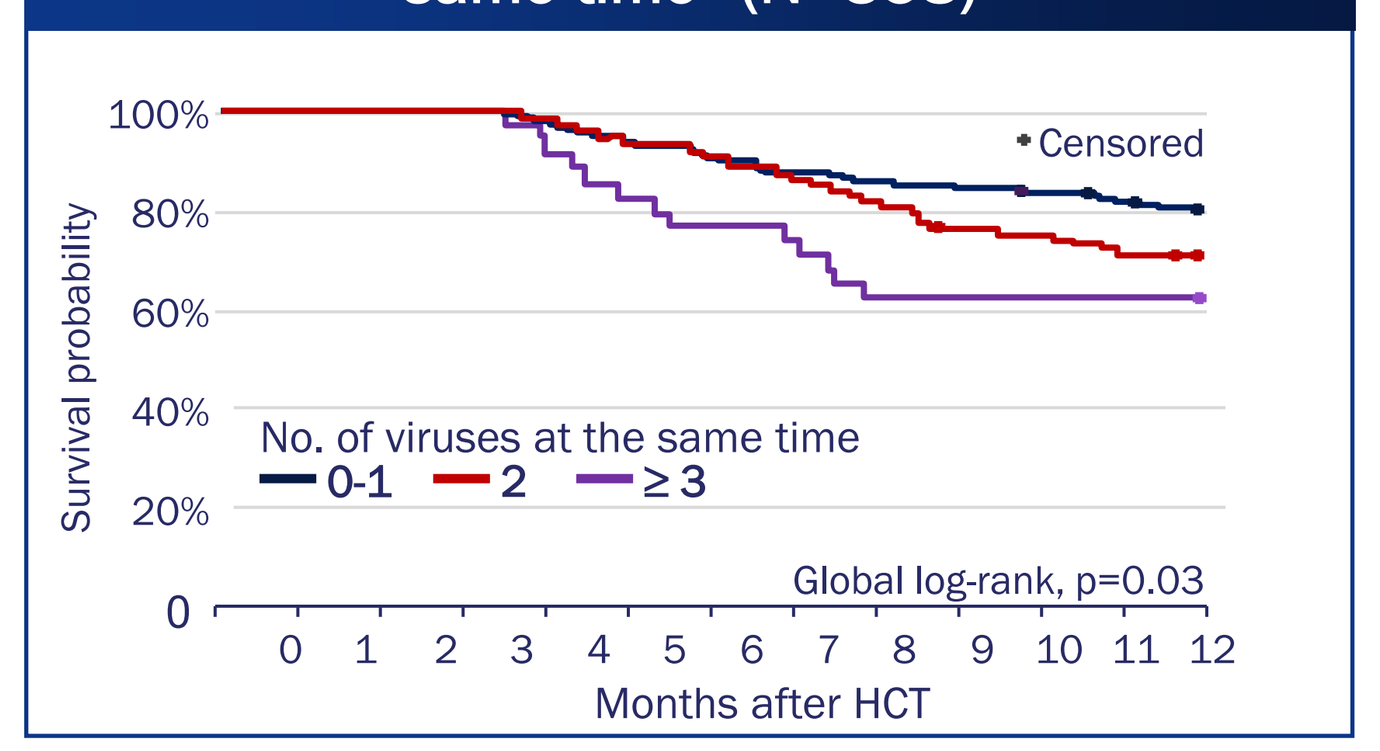
Survival probability
100%
80%
60%
40%
20%
0
0 1 2 3 4 5 6 7 8 9 10 11 12
Months after HCT
Censored
No. of viruses at the same time
0-1
2
≥ 3
Global log-rank, p=0.03

- 61% of patients were treated with antiviral therapies, excluding rituximab, within the first 100 days

Approximately 70% Incidence of Clinically Significant Infection and Disease due to AdV, BKV, CMV, EBV, HHV-6, or JCV in High-Risk Allo-HCT Patients

Estimated incidence of clinically significant viral infection or disease

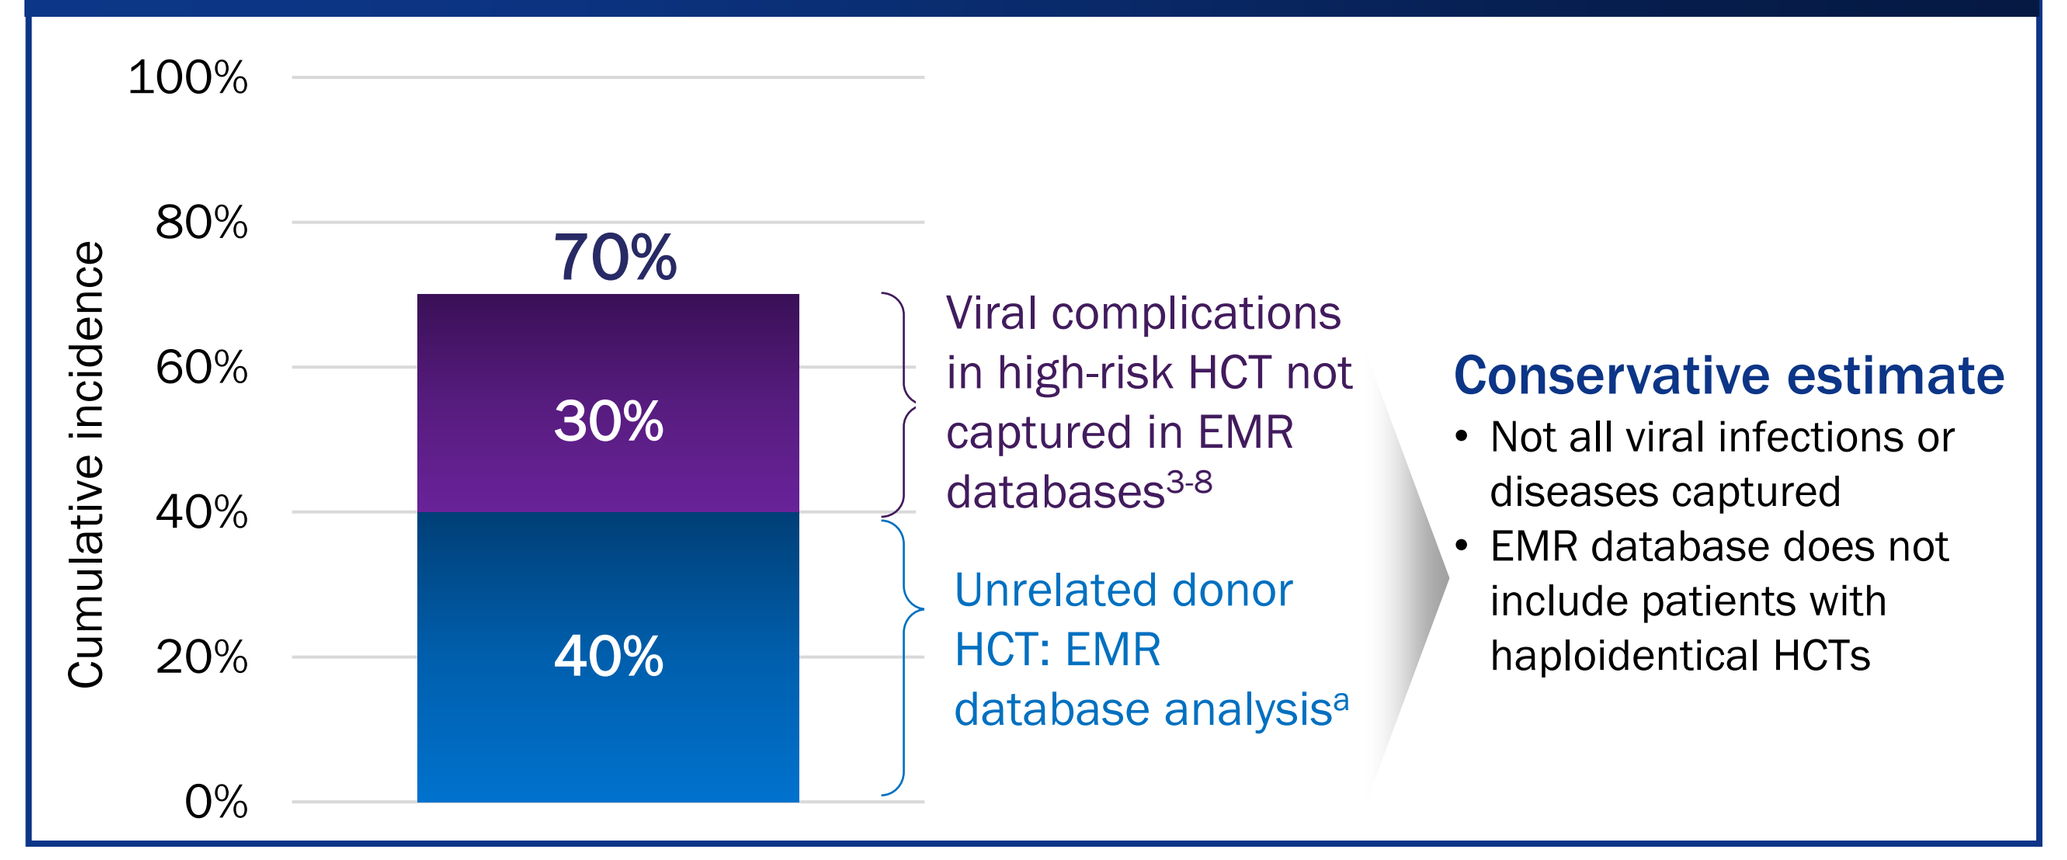
Cumulative incidence
100%
80%
60%
40%
20%
0%
70%
30%
40%
Viral complications in high-risk HCT not captured in EMR databases3-8
Unrelated donor HCT: EMR database analysisa
Conservative estimate
• Not all viral infections or diseases captured
• EMR database does not include patients with haploidentical HCTs

[a]>1400 patients identified between Jan 2018 and Apr 2021 through use of ganciclovir, valganciclovir, foscarnet, cidofovir, or rituximab or ICD-9/10 code for viral disease where available (e.g., does not include virus associated hemorrhagic cystitis)

- Allogeneic HCT(allo-HCT) patients are at high risk for common dsDNA infections: AdV, CMV, BKV, EBV, HHV-6 and JCV
- Allo-HCT patients at highest risk: haploidentical donor, UCB, MMUD, MUD, MMRD, T cell depletion (Approx. 75% of total allo-HCT patients[2])

There is an unmet need for preventive therapies targeting multiple viruses in high-risk allo-HCT patients

Posoleucel (ALVR105)

- Allogeneic, off-the-shelf, multivirus-specific T-cell (VST) therapy, targeting AdV, BKV, CMV, EBV, HHV-6 or JCV[a]
- 93% response rate in Phase 2 CHARMS study[9]

Phase 2 multi-virus prevention open label study design[a]

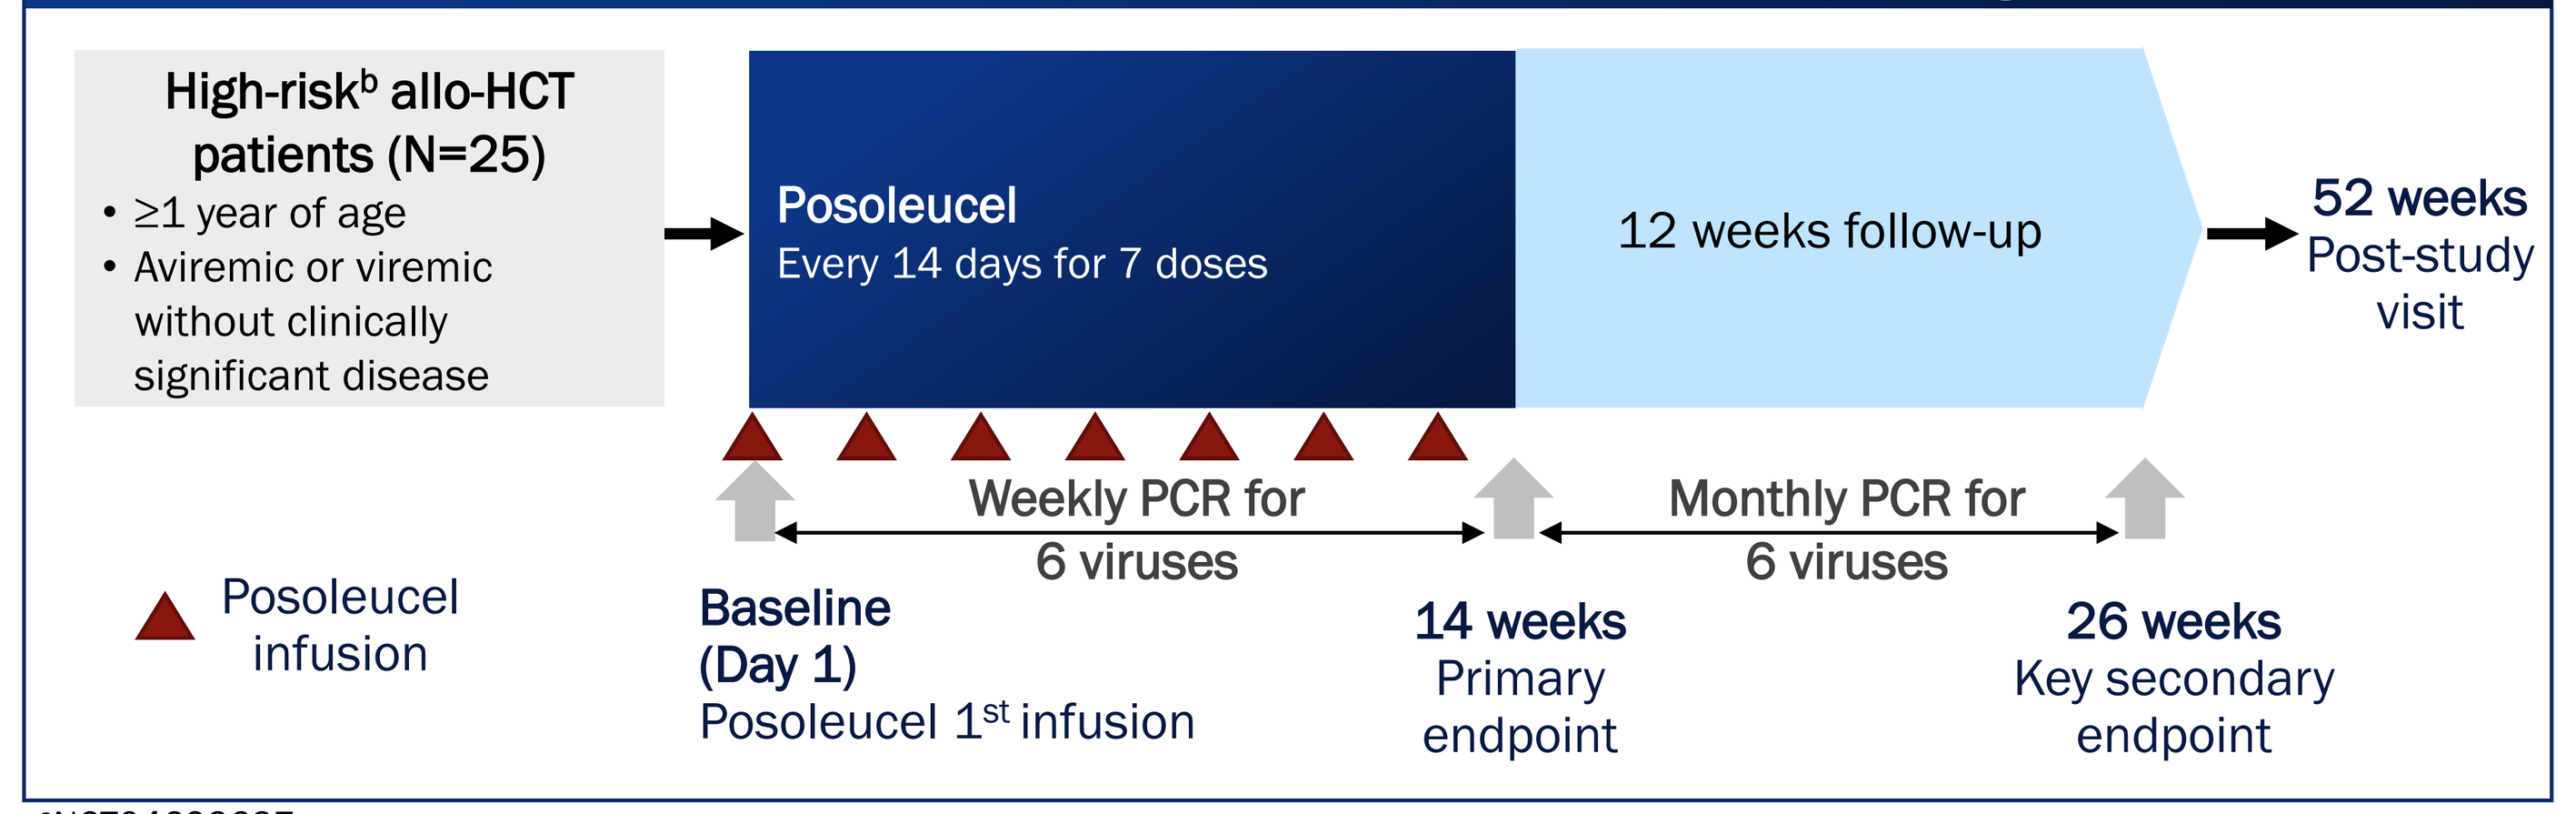
High-riskb allo-HCT patients (N=25)
• ≥1 year of age
• Aviremic or viremic without clinically significant disease
Posoleucel
Every 14 days for 7 doses
12 weeks follow-up
52 weeks
Post-study visit
Weekly PCR for 6 viruses
Monthly PCR for 6 viruses
Posoleucel infusion
Baseline (Day 1)
Posoleucel 1st infusion
14 weeks
Primary endpoint
26 weeks
Key secondary endpoint

[a]NCT04693637
[b]High-risk allo-HSCT defined as: umbilical cord donor, haploidentical donor, MMRD, MUD, MMUD, recipient of T-cell depletion (ex vivo, alemtuzumab, ATG), persistent lymphopenia <180/mm³

Primary endpoint: The number of new onset clinically significant infections or episodes of end-organ disease through Week 14

Clinically significant viral infection	1. Above viral load threshold • CMV: >910 IU/mL • EBV/AdV: >10,000 copies/mL OR >1,000 copies/mL and rising[a] AND 2. Initiation of preemptive antiviral therapy
End-organ disease	Signs or symptoms of organ damage from AdV, BKV, CMV, EBV, HHV-6, or JCV

[a]Defined as two consecutive results of >1,000 copies/mL with the second result being higher than the first and drawn at least 48 hours after the first

Results

Baseline Demographics

Patient Characteristics	N=23
Age, median years (range)	59 (14-73)
Female, n (%)	12 (52%)
Non-Caucasian or Latino, n (%)	11 (48%)
Diagnosis, n (%)	
Leukemia	14 (61%)
Myelodysplasia/Myelofibrosis	3 (13%)
Lymphoma	2 (9%)
Sickle cell anemia	2 (9%)
Other[a]	2 (9%)
Donor type, n (%)	
Haploidentical	14 (61%)
Mismatched Unrelated	6 (26%)
Matched Unrelated[b]	2 (9%)
Umbilical Cord Blood	1 (4%)
Myeloablative conditioning, n (%)	12 (52%)
Baseline viremia[c], n (%)	10 (43%)
Letermovir prophylaxis, n (%)	14 (61%)

[a]Multiple myeloma and adrenoleukodystrophy
[b]Matched unrelated transplant recipients included if also met another high-risk criterion: T cell depletion or persistent lymphopenia
[c]1 AdV, 7 BKV, 2 EBV and/or 4 HHV-6 viremia(s) detected in 10 patients

Study Disposition

- 97% (32/33) of screened patients with matching posoleucel cell lines
- Median 3/8 HLA allele matches
- Median 5 posoleucel doses to date
- Posoleucel administered a median of 42 days post allo-HCT

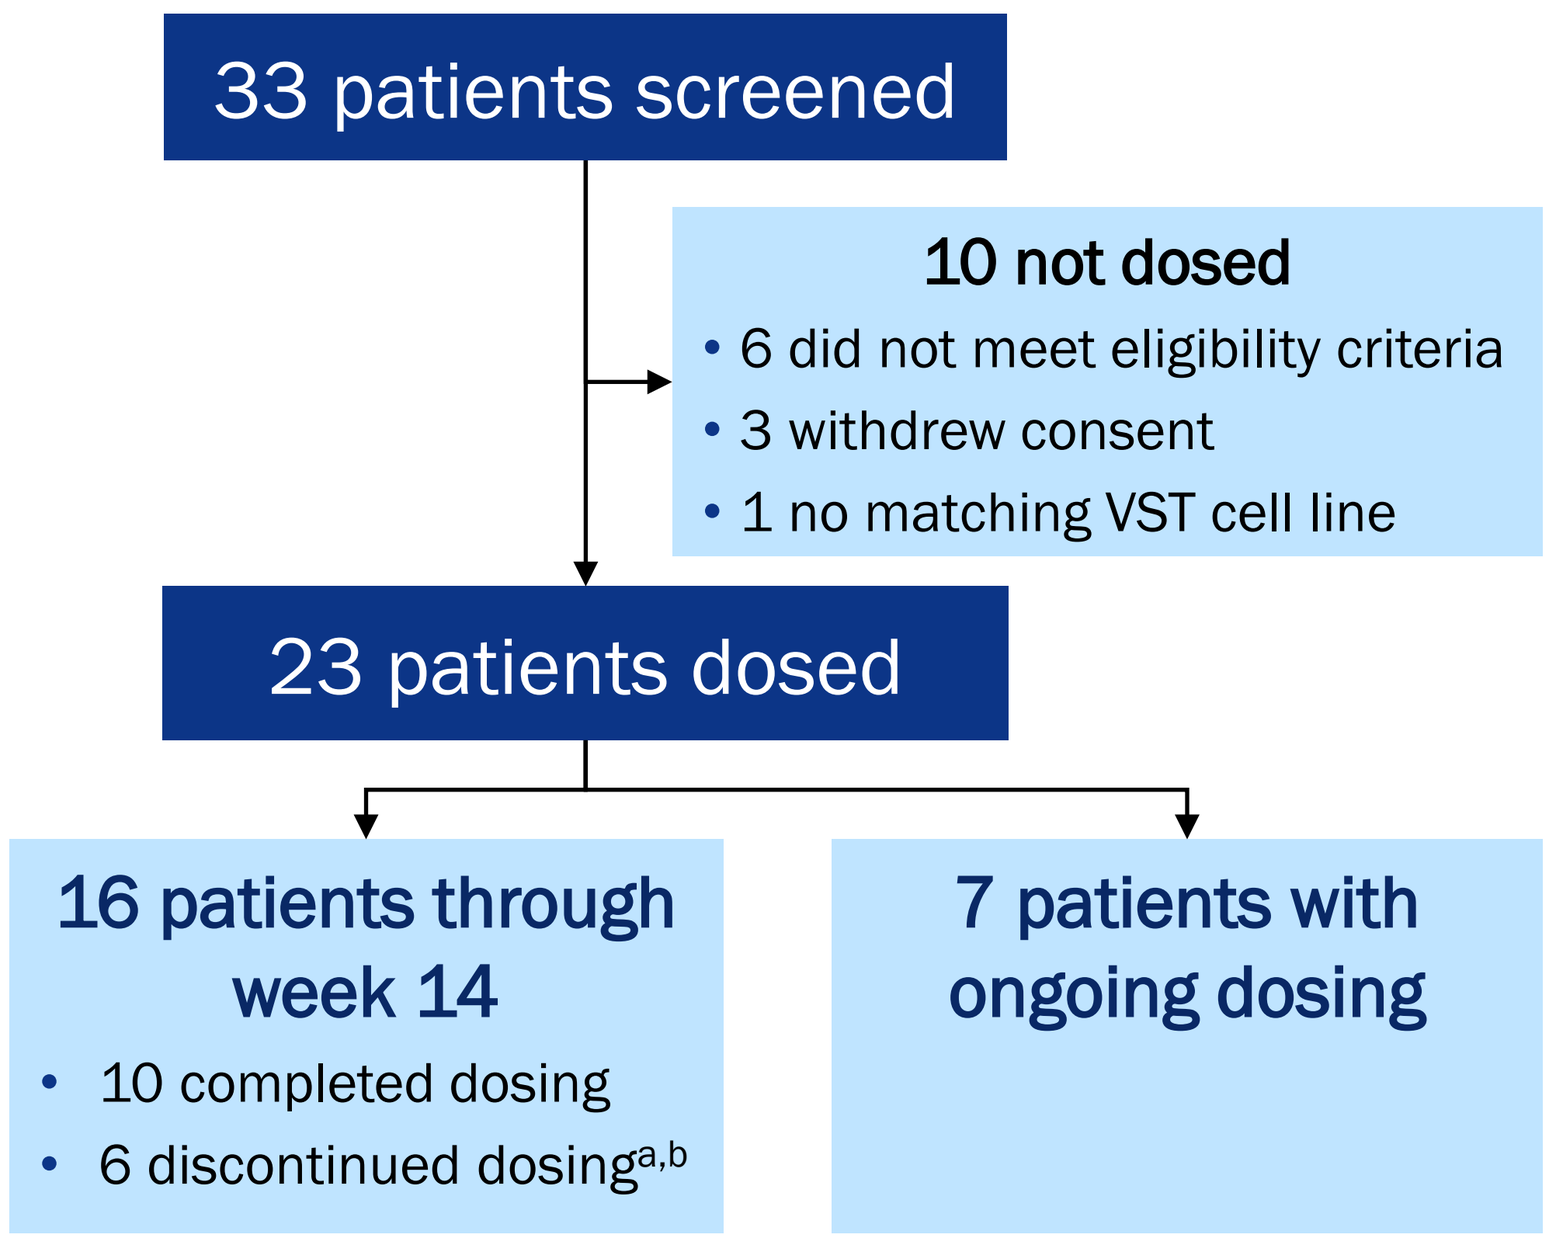
33 patients screened
10 not dosed
• 6 did not meet eligibility criteria
• 3 withdrew consent
• 1 no matching VST cell line
23 patients dosed
16 patients through week 14
• 10 completed dosing
• 6 discontinued dosinga,b
7 patients with ongoing dosing

[a]4 patients due to AEs assessed not related to posoleucel; 1 patient due to AEs assessed as possibly related to posoleucel; 1 patient withdrew consent
[b]Median (range) posoleucel doses: 3 (1-6)

Safety and Tolerability

Events	N=23
Common adverse events (AEs)	
Diarrhea	7 (30%)
Weight decrease	5 (22%)
SAEs	11 (48%)
Treatment related SAE	2 (9%)[a]
Deaths	1 (4%)[b]
Posoleucel DC due to TEAEs	3 (13%)[c]

Events	N=23
Adverse events of interest	
Acute GVHD II-IV	6 (26%)
Grade II	2 (9%)
Grade III	4 (17%)
Grade IV	0 (0%)
Cytokine release syndrome	0 (0%)
Infusion reaction	1 (4%)[d]

[a]1 patient with infusion reaction; 1 patient with acute GVHD; [b]Relapse of primary malignancy ~2 months after the 7th dose of posoleucel; [c]2 patients assessed as not related to posoleucel; 1 patient assessed as possibly related to posoleucel; [d]Tolerated subsequent posoleucel doses with pre-medication (diphenhydramine).

- No unexpected treatment-emergent adverse events (TEAEs) or serious adverse events (SAEs) were reported
- 6/23 (26%) Grade II-IV acute GVHD
 - Consistent with 35-50% of Grade II-IV GVHD reported in high risk allo-HCTs[11-13]
 - No association between GVHD and # of HLA matches
 - No association between HD and # of doses

Preliminary Result: Primary Endpoint (week 14)

- **3 clinically significant infections**
 - 2 patient started on pre-emptive valganciclovir for CMV post withdrawal of letermovir
 - 1 patient started rituximab for EBV in the setting of high dose steroids
- **No end organ disease[a]**

% Clinically significant infections through week 14

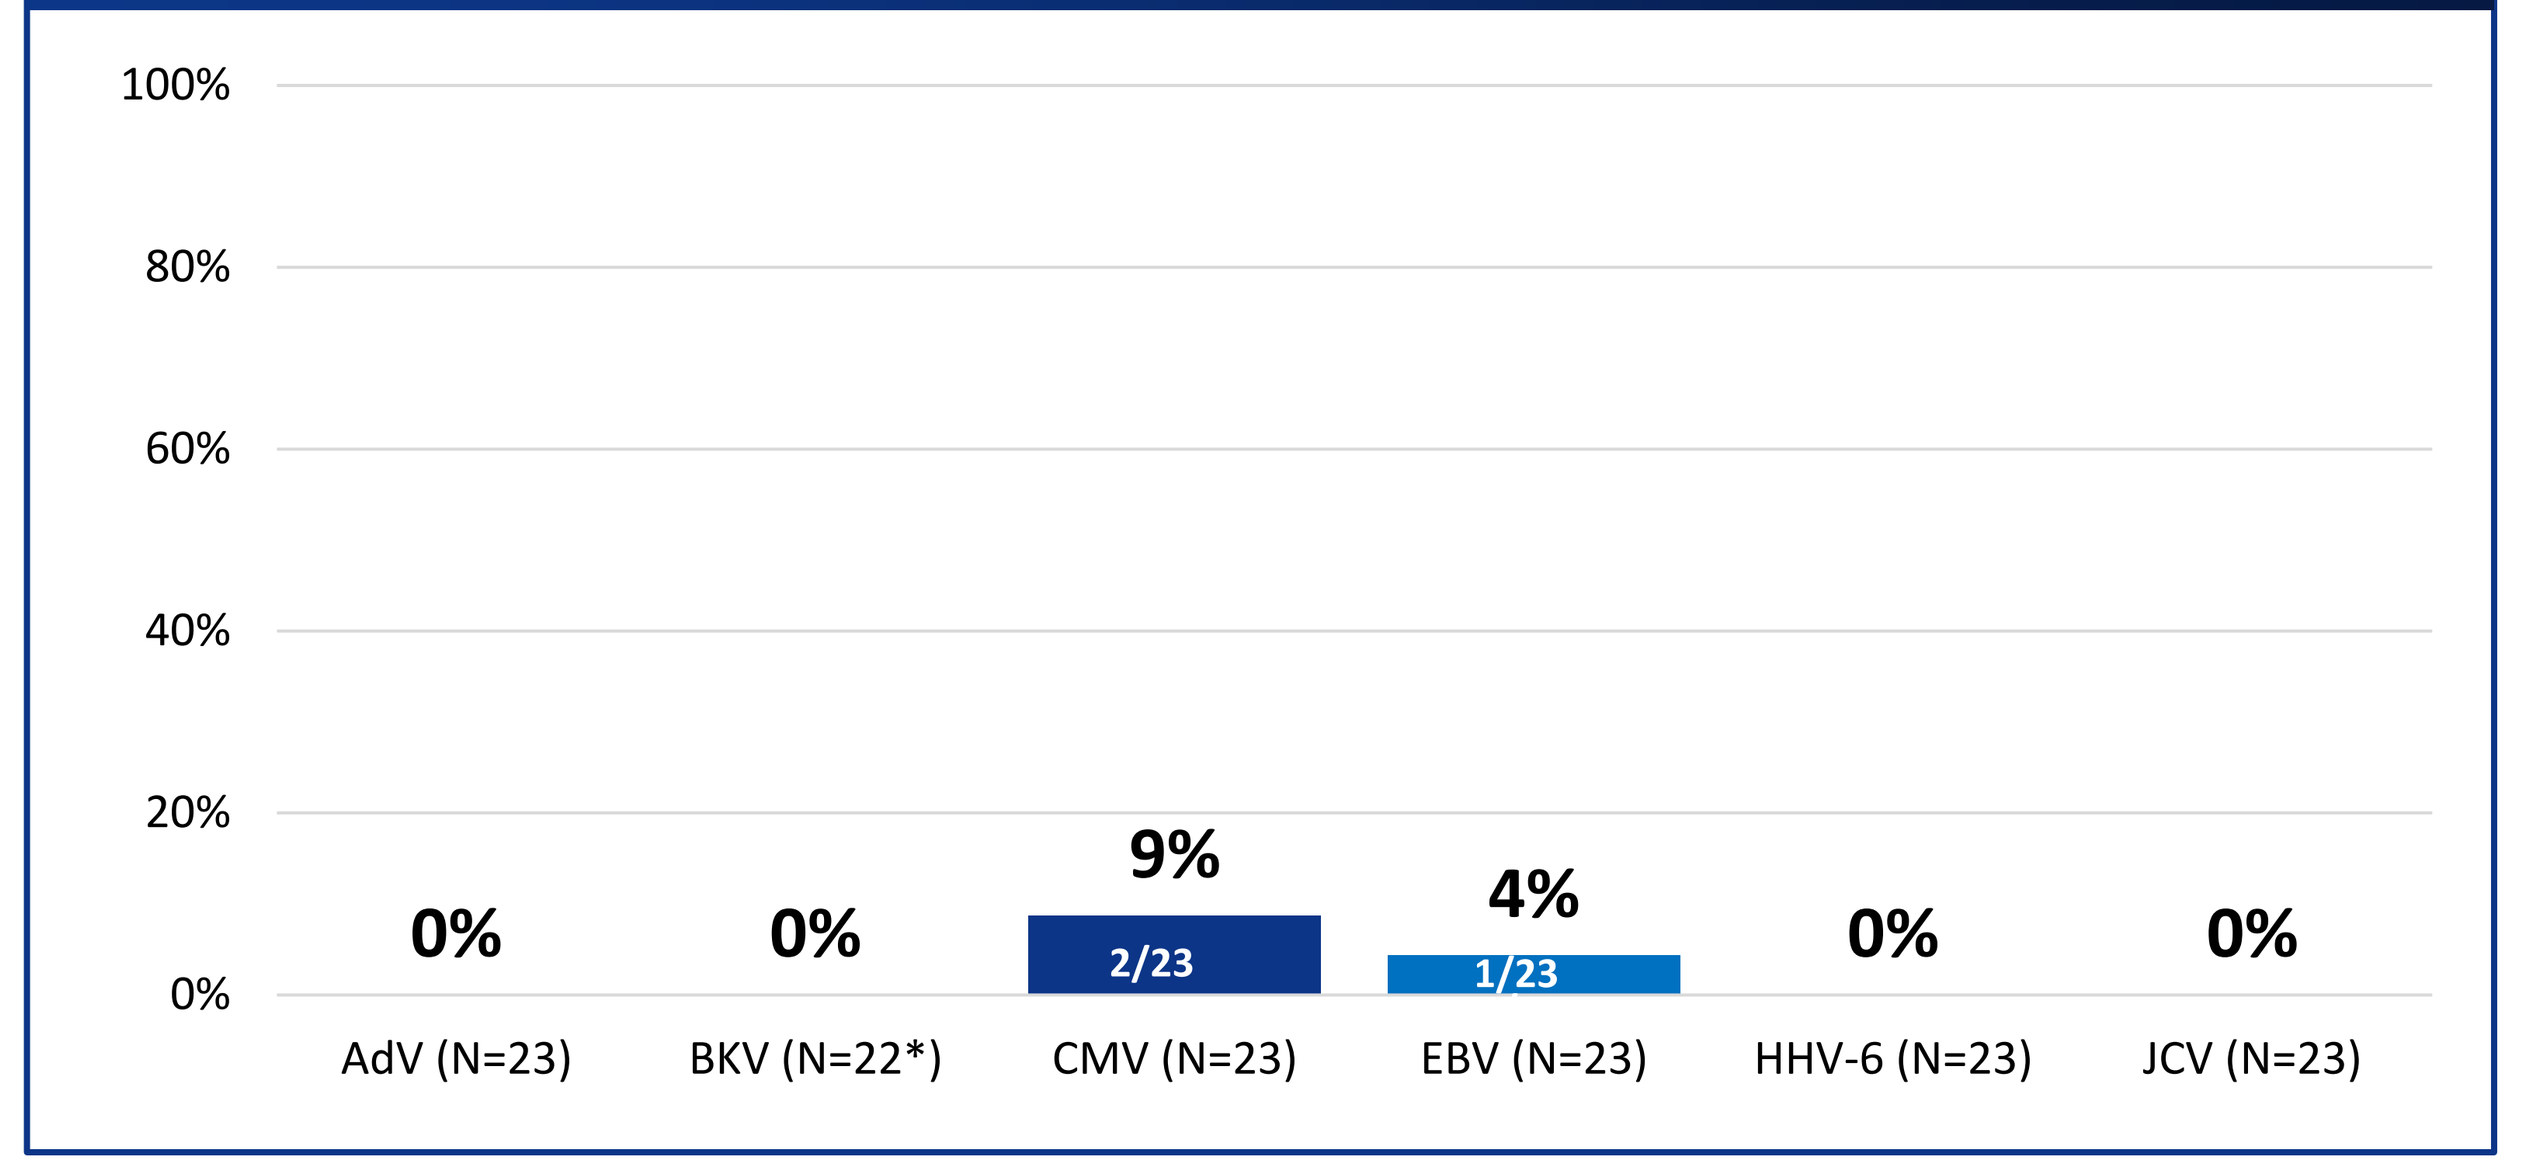
100%
80%
60%
40%
20%
0%
0%
0%
9%
2/23
4%
1/23
0%
0%
AdV (N=23)
BKV (N=22*)
CMV (N=23)
EBV (N=23)
HHV-6 (N=23)
JCV (N=23)

[a]One patient excluded due to BKV hemorrhagic cystitis at baseline

Preliminary Result: Secondary Endpoint (week 26)

- **2 new clinically significant infections**
 - 1 patient started on pre-emptive valganciclovir for CMV post withdrawal of letermovir
 - 1 patient started cidofovir for AdV in the setting of high dose steroids
- **No end organ disease**

Incidence of clinically significant infections by 6 target viruses in high-risk allo-HCT patients receiving posoleucel

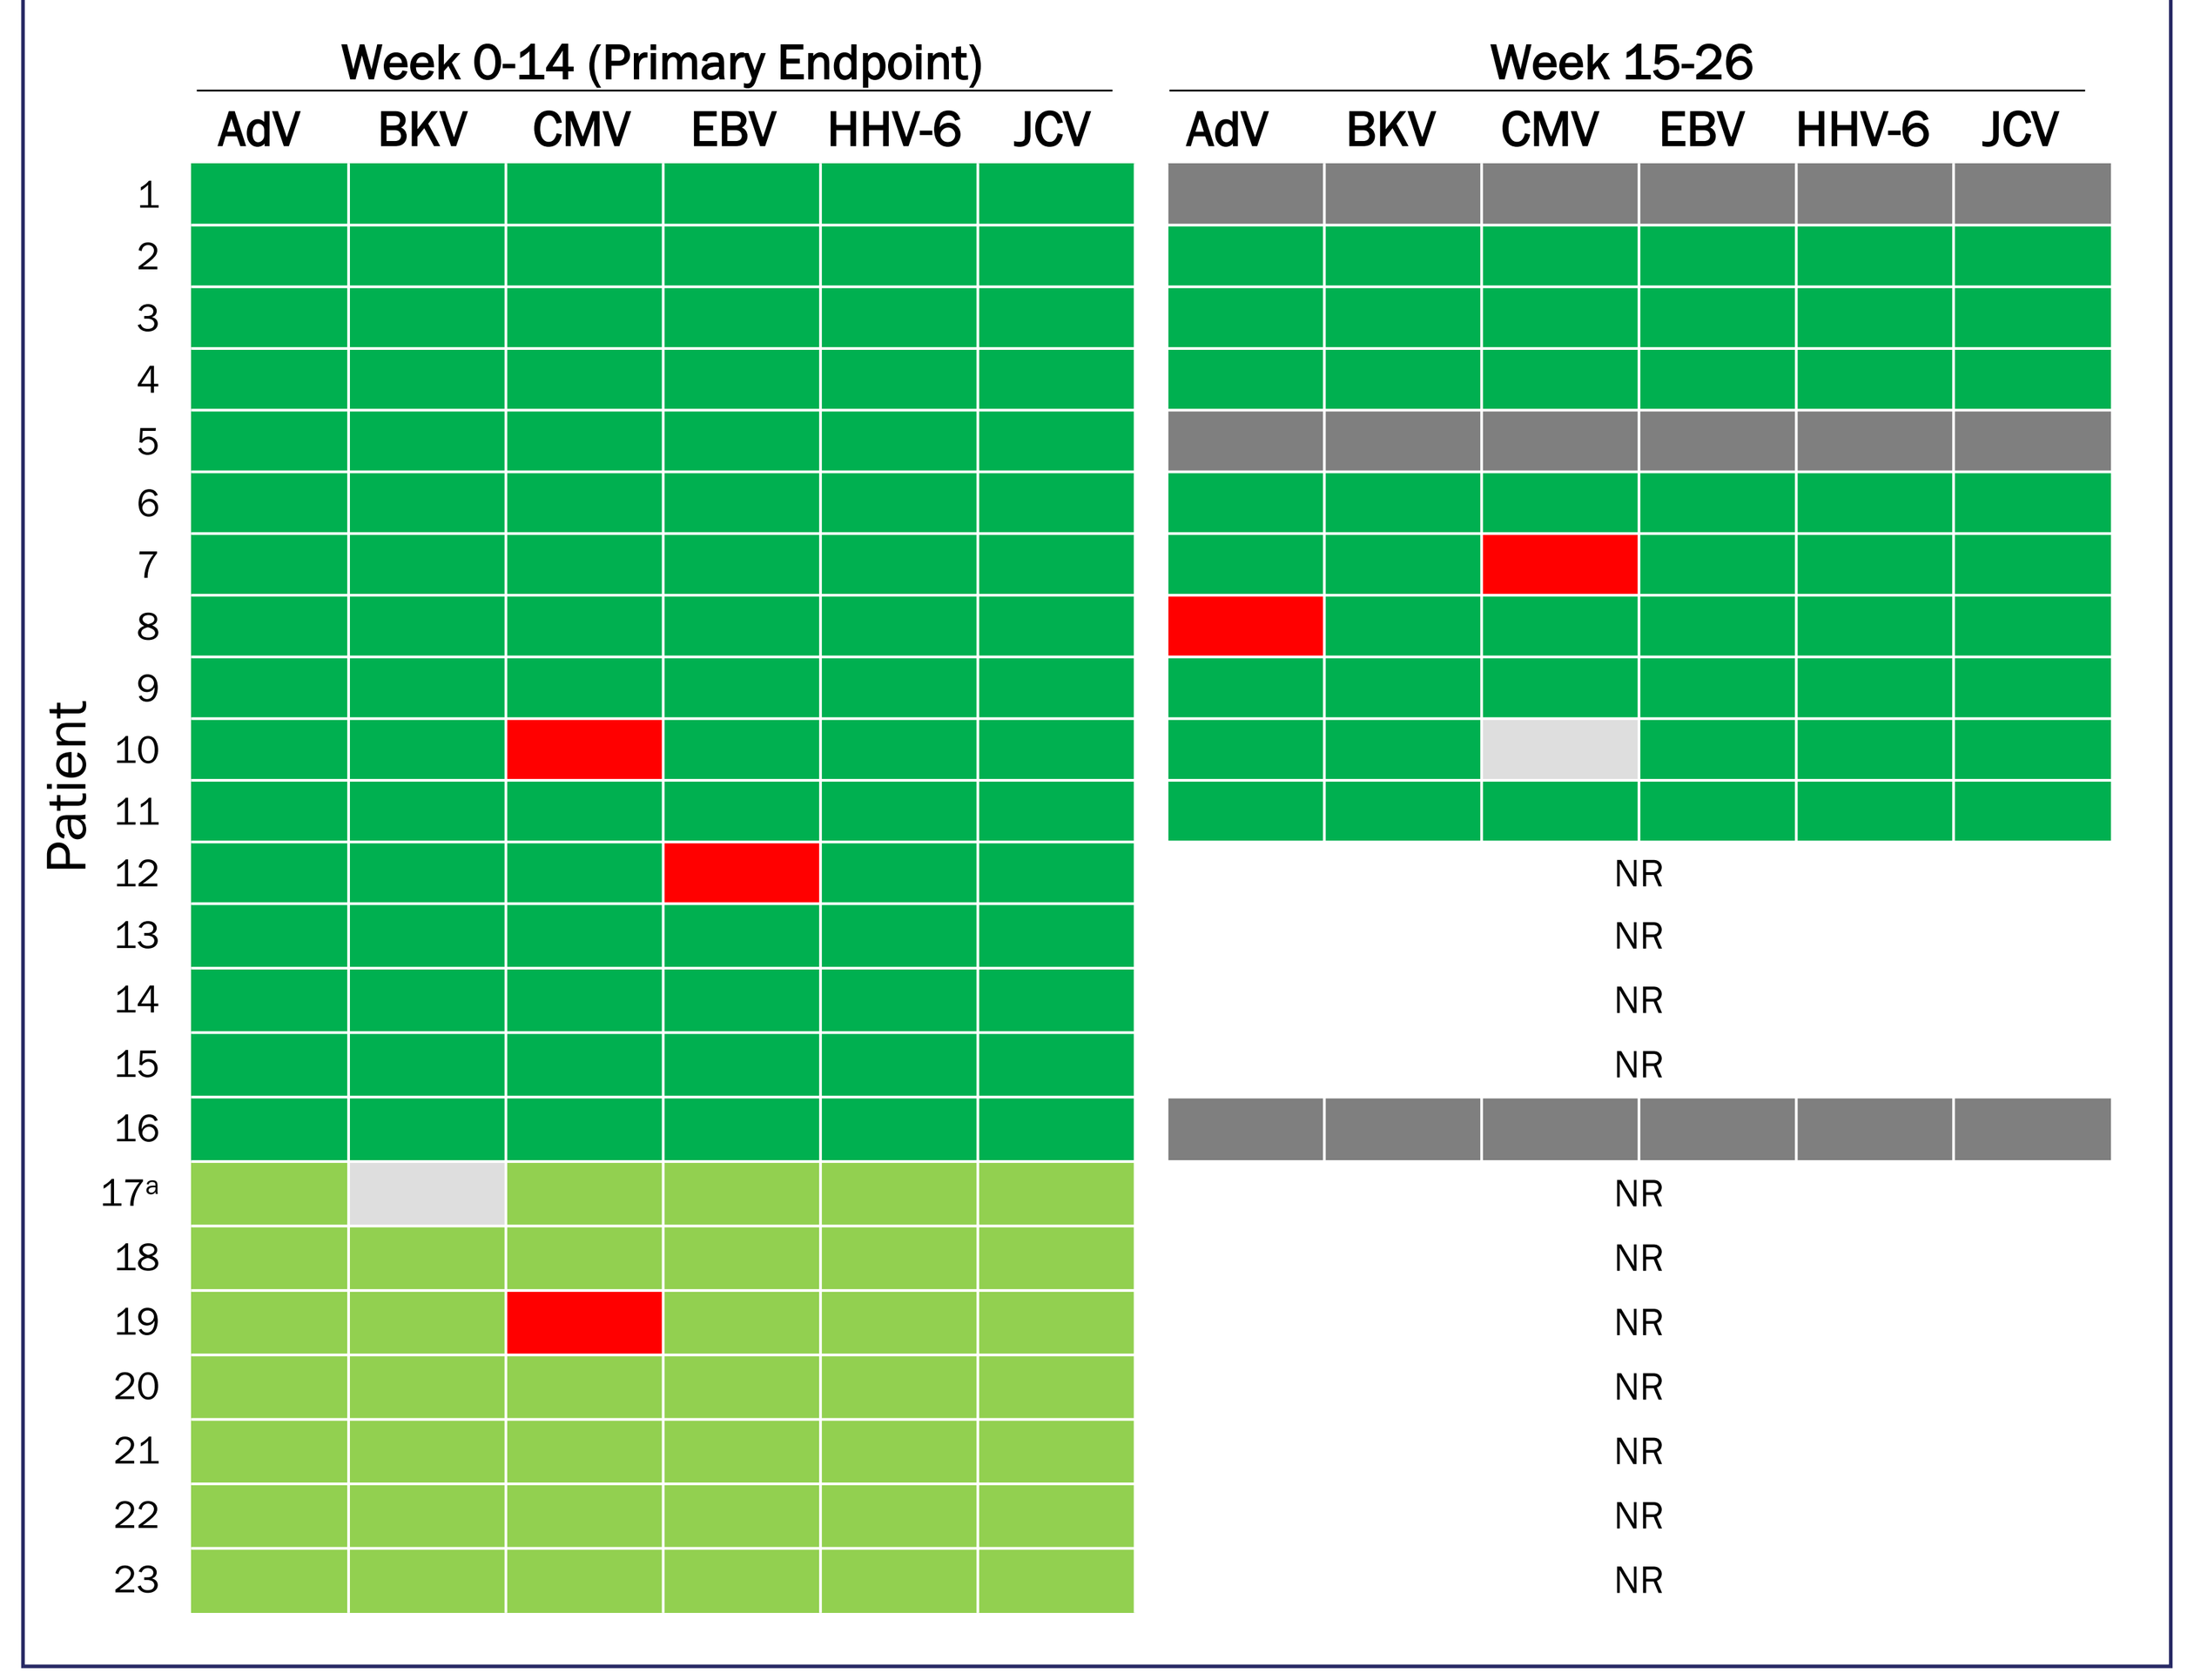
Week 0-14 (Primary Endpoint)
Week 15-26
AdV BKV CMV EBV HHV-6 JCV
AdV BKV CMV EBV HHV-6 JCV
Patient
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17a 18 19 20 21 22 23
NR

- Patients with completed or stopped dosing who did not develop clinically significant infection or disease
- Patients with ongoing dosing/monitoring who have not developed clinically significant infection or disease
- Patients who developed newly onset clinically significant infection or disease
- Patients sample not collected
- Previous clinically significant infection or disease
- NR Week 18 Timepoint not yet reached

[a]BKV hemorrhagic cystitis at baseline

CMV Infection Controlled without Letermovir or Pre-emptive Therapy while on Posoleucel

- 61YO / Cutaneous T cell lymphoma / Myeloablative conditioning / MMUD HCT
- CMV serostatus: D-/R+
- Last dose of letermovir was 1 day prior to 1st posoleucel dose
- No pre-emptive therapy during study

CMV viral load in a patient receiving posoleucel

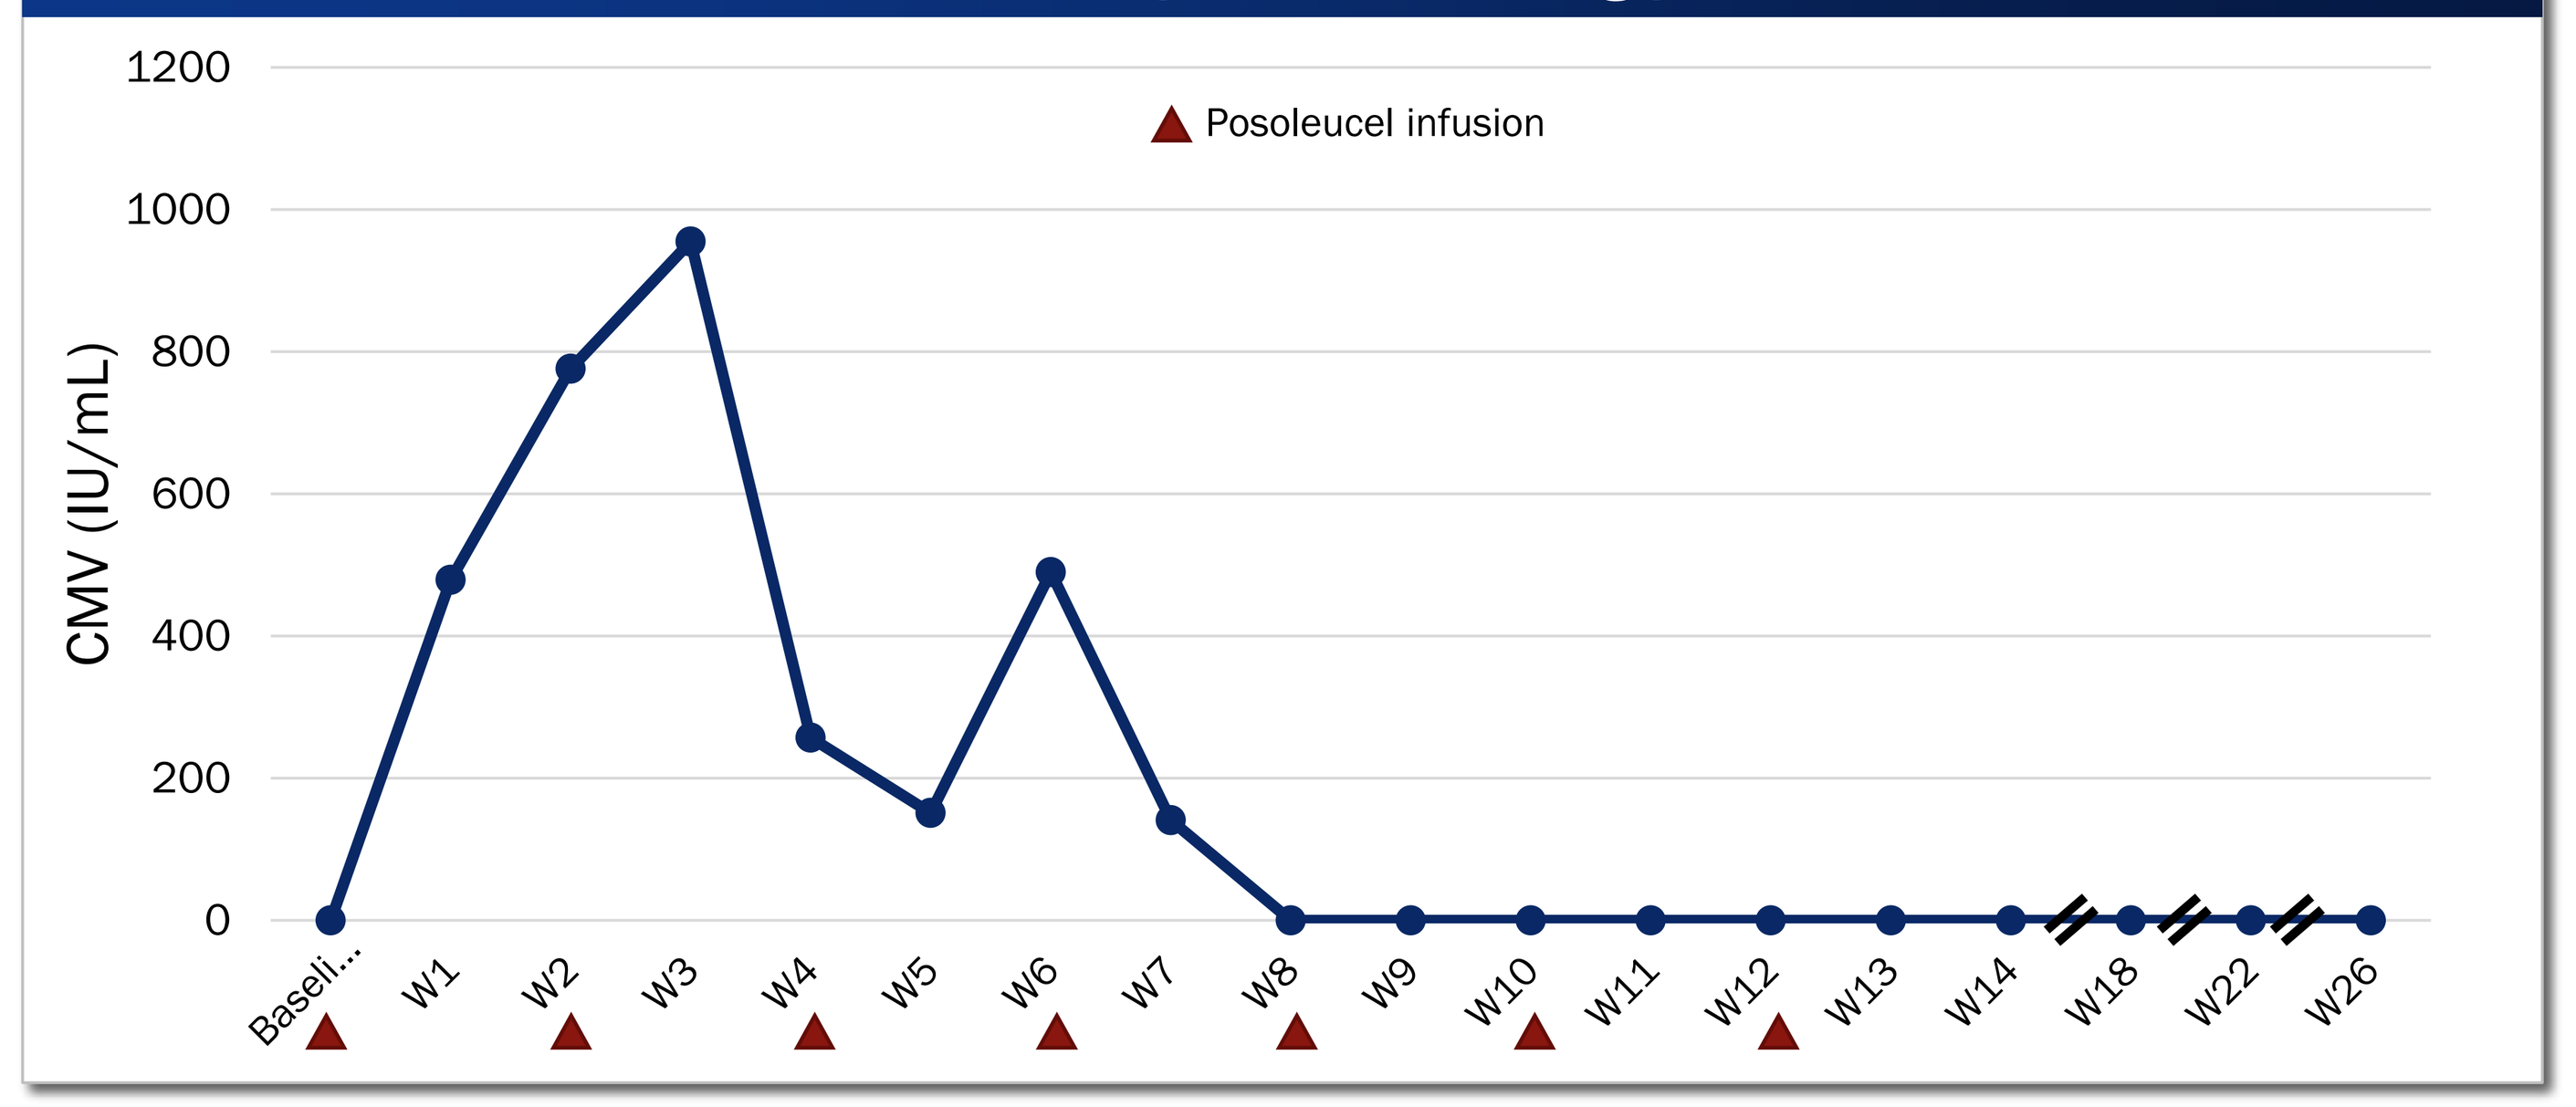
CMV (IU/mL)
1200
1000
800
600
400
200
0
Posoleucel infusion
Baseli...
W1 W2 W3 W4 W5 W6 W7 W8 W9 W10 W11 W12 W13 W14 W18 W22 W26

Biomarker Summary

- Preliminary assessment of T cell activity by IFNγ ELIspot (N=4) shows increased T cell activity against the infecting target viruses in patients with clinically significant infection
- Preliminary data from TCR sequencing (N=6) demonstrate posoleucel can be detected throughout the 14-week dosing period

Conclusions

- Approximately 70% of high-risk allo-HCT patients develop clinically significant viral infections or disease due to AdV, BKV, CMV, EBV, HHV-6, or JCV
- In the ongoing, open-label cohort of the Phase 2 Multi-Virus Prevention study:
 - **High-risk allo-HCT patients receiving posoleucel had low rates of clinically significant viral infections and no end-organ disease**
 - **Repeat dosing of posoleucel was generally safe and well tolerated**
- These results support the evaluation of posoleucel for the prevention of infections and diseases from the 6 targeted viruses in the upcoming randomized, placebo-controlled Phase 3 trial

Acknowledgement

- We extend our thanks to the patients, their families, and all participating investigators
- This study is funded by AlloVir

References

1. Hill et al, *Blood* 2017; **2.** CIBMTR 2020 summary report; **3.** Slade et al. *Transpl Infect Dis*. 2017; **4.** Mohty et al. *British Journal of Haematology* 2019; **5.** Salamonowicz-Bodzioch et al. *Ann Hematol*. 2021; **6.** Mojtaba et al. *Biol Blood Marrow Transplant.* 2019; **7.** El-Zimaity et al. *Blood* 2014; **8.** Gargiulo et al. *eCancer* 2014; **9.** Tzannou et al. *Blood* 2020; **10. 11.** Malki et al. *Blood Adv*. 2021; **12.** Saliba RM, et al. Abstract 31. Presented at: TCT 2020; **13.** Chen et al., *Bone Marrow Transplant.* 2017.

Disclosures

SD reports relationship with Merck, AlloVir, Astellas, Asepticope, and Sire/Takeda. MS reports relationship with Takeda, MorphSys, Astellas, Intellisphere, AbbVie, Pharmacyclics, AlloVir, Janssen, Epizyme, Bristol Myers Squibb, Beigene, Asctinium, Rafael, GSK, Incyte, Seattle Genetics, Novartis, Genetech, Amgen, and Celgene. GD reports relationship with Novartis, AlloVir, and Eliana. KB, MW, and ES report relationship with AlloVir. TT reports no relationship. JH reports relationship with Gilead, Amplyx, Allovir, Allogene therpaeutics, CRISPR therapeutics, CLS Behring, OptumHealth, Octapharm, Takeda, and Karius.